Cost-Effectiveness of Darunavir for the Management of HIV-Infected, Treatment-Experienced Adults in Canada

J Mauskopf,[1] A Brogan,[1] C Malmberg,[2] P Hwang[2]

[1]RTI Health Solutions, Research Triangle Park, NC, USA; [2]Janssen-Ortho Inc., Toronto, ON, Canada

BACKGROUND

- The introduction of protease inhibitors (PIs) in the mid-1990s represented a major advance in the treatment of HIV infection. It has resulted in sustained viral suppression, improved immunologic function, and marked reduction in morbidity and mortality rates.
- However, current treatment with PIs is limited by factors such as adverse effects, drug interactions, and the development of resistance.
- Darunavir (Prezista®, TMC114) is a novel PI with demonstrated superior efficacy to currently available PIs for the treatment of HIV infection in treatment-experienced adults who have failed prior antiretroviral therapy.
- An understanding of the value for money of darunavir compared to currently available PIs is required by health care decision makers to identify darunavir's appropriate place in therapy.

OBJECTIVE

To evaluate the cost-effectiveness, from a Canadian provincial Ministry of Health perspective, of ritonavir-boosted darunavir (darunavir/r) plus an optimized background regimen (OBR) compared to currently available PIs plus OBR.

The population of interest for this analysis is people with HIV infection who have previously failed antiretroviral therapy and who are starting a new, multi-drug antiretroviral regimen that includes PIs plus an OBR made up of nucleoside reverse transcriptase inhibitors with or without enfuvirtide.

METHODS

Model Treatment Pathways

- Figure 1 illustrates the treatment pathways compared in this economic evaluation.
- After starting each new treatment regimen, the model allowed three sequential stages of CD4+ cell-count change:
 - Period of rapidly increasing CD4+ cell count,
 - Period of slowly increasing or stable CD4+ cell count, and
 - Period of declining CD4+ cell count until switch to new therapy regimen or death.

Figure 1. Model Treatment Pathways

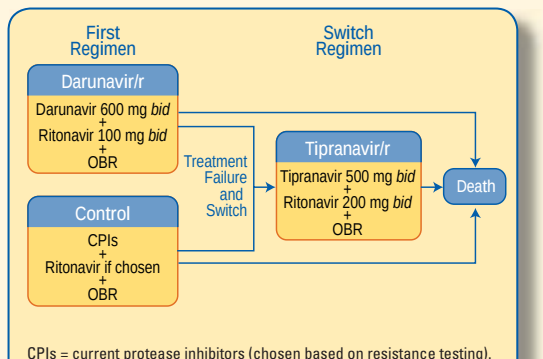
First Regimen
Switch Regimen
Darunavir/r
Darunavir 600 mg bid + Ritonavir 100 mg bid + OBR
Control
CPIs + Ritonavir if chosen + OBR
Treatment Failure and Switch
Tipranavir/r
Tipranavir 500 mg bid + Ritonavir 200 mg bid + OBR
Death

CPIs = current protease inhibitors (chosen based on resistance testing).
OBR = optimized background regimen
Treatment failure defined as a decline in CD4+ cell count. Switch may occur several time periods after start of CD4+ cell count decline.

Markov Model Structure and Input Parameters

- A Markov model with a 3-month cycle period was developed to follow a treatment-experienced HIV cohort through six possible health states, defined by CD4+ cell-count ranges (0-50, 51-100, 101-200, 201-350, 351-500, and >500 cells/mm^3), and eventually to the death state.
- Transition probabilities between the Markov model health states were calculated from the POWER 1 and POWER 2 clinical trial results for the darunavir/r and control regimens and from the RESIST 1 and RESIST 2 clinical trial results for the tipranavir/r switch regimen and from other published sources.
- Clinical trial data used to compute the transition probabilities included the proportion of individuals with different levels of virologic response to treatment at 24 weeks and the changes in CD4+ cell count at 24 and 48 weeks associated with the different virologic response groups for each treatment option (Tables 1-3).
- Antiretroviral drug costs were based on usage rates in the clinical trials, and the mean daily cost for each drug was computed using the recommended dose in U. S. DHHS guidelines. Unit costs were obtained from the Ontario and Quebec formularies. The total daily cost of PIs is $30.52 for the darunavir/r regimen and $38.36 for the tipranavir/r regimen.
- Other costs and utility data were estimated based on published Canadian sources. HIV-related and non-–HIV-related mortality were taken from published studies and Canadian national statistics, respectively (Table 4).
- All costs were estimated in 2006 Canadian dollars and both costs and outcomes were discounted at 5%.
- Extensive sensitivity and variability analyses were performed to test the robustness of the cost-effectiveness results.

Table 1. Virologic Response Rates at 24 Weeks

Treatment Regimen	<50 Copies/mL	≥1 Log_{10} Drop, >50 Copies/mL	<1 Log_{10} Drop
Darunavir/r	45.0%	25.2%	29.8%
Control	12.1%	8.9%	79.0%
Tipranavir/r	23.9%	17.3%	58.8%

Sources: Pooled data from POWER 1 and POWER 2 clinical trials, Janssen-Ortho data on file, for darunavir/r and control; and from Cooper et al., 2005; Hicks et al., 2004; and Cahn et al., 2004 for tipranavir/r.

Table 2. Estimated 3-Month Initial Increase in CD4+ Cell Count by 24-Week Virologic Response: First and Switch Regimens

Treatment Regimen	<50 Copies/mL Mean (SD)	≥1 Log_{10} Drop, >50 Copies/mL Mean (SD)	<1 Log_{10} Drop Mean (SD)
Darunavir/r	54.19 (55.94)	73.76 (73.10)	24.38 (50.77)
Control	26.69 (53.52)	32.18 (39.93)	4.22 (54.83)
Tipranavir/r*	24.76 (25.56)	33.70 (33.41)	11.14 (23.21)

*The CD4+ cell count increases by 24-week virologic response for tipranavir/r were imputed based on the published data on the mean value of CD4+ cell-count increase and the proportion of trial participants in each virologic response category, assuming values proportionate to those observed for the darunavir/r arm of the POWER 1 and POWER 2 clinical trials.

Sources: Pooled data from POWER 1 and POWER 2 clinical trials, and data from RESIST trials presented in Katlama et al., 2006; Cahn et al., 2004; and Hicks et al., 2004.

Table 3. Durations of CD4+ Cell-Count Changes by 24-Week Virologic Response: First and Switch Regimens

	<50 Copies/mL	≥1 Log_{10} Drop, >50 Copies/mL	<1 Log_{10} Drop
1. Initial CD4+ cell-count increase			
Darunavir/r	0.5 years	0.5 years	0.5 years
Control	1 year	0.5 years	0.5 years
Tipranavir/r	1 year	0.5 years	0.5 years
2. Stable or slowly increasing CD4+ cell count			
Darunavir/r	2 years	0.5 years	0 years
Control	1.5 years	0.5 years	0 years
Tipranavir/r	1.5 years	0.5 years	0 years
3. Declining CD4+ cell count before switching or stopping regimen			
Darunavir/r	3 years	3 years	1 year
Control	3 years	3 years	1 year
Tipranavir/r	Remaining lifetime	Remaining lifetime	Remaining lifetime

Sources: Janssen-Ortho Inc data on file, 2006; Tarwater et al., 2001; Kaufmann et al., 2003; Hunt et al., 2003; Smith et al., 2003; Garcia et al., 2004; Deeks et al., 2002; Ledergerber et al., 2004.

Table 4. Utility Values, HIV-Related Mortality Rates, and Annual Costs for Resources Other Than ARV Drugs, by CD4+ Cell-Count Range

CD4+ Cell-Count Range (Cells/mm3)	Utility Value	Annual Risk of HIV-Related Death (%)	Annual Costs
>500	0.95	0.4%	$2,779
351 – 500	0.93	0.4%	$3,291
201 – 350	0.93	0.8%	$4,242
101 – 200	0.85	2.2%	$6,327
50 – 100	0.85	5.5%	$6,327
<50	0.78	17.6%	$14,138

Sources: Utility values, Simpson et al., 2004; HIV-related mortality rates: Mocroft et al., 2003; annual costs for inpatient, outpatient, and emergency department resources and medications other than ARV drugs: McMurchy et al., 1998; Krentz et al., 2003, inflated to 2006 Canadian dollars using inflation rates from Statistics Canada 2006.

RESULTS

Table 5. One-Year Cost-Effectiveness Analysis for Darunavir/r Compared to the Control (Standard of Care) Regimen

Outcome Measure	Darunavir/r	Control	Difference
One-year cost	$37,190	$33,627	$3,563
Probability of viral load <50 copies/mL at 48 weeks	0.46	0.10	0.36
Incremental cost per additional person with a viral load of <50 copies/mL			$9,897

Table 6. Lifetime Cost-Utility Analysis of Darunavir/r Compared to Control: Base Case, Discounted at 5%

Outcome Measure	Darunavir/r	Control	Difference
Life-years	9.02	7.77	1.26
QALYs	8.05	6.78	1.27
Lifetime costs	$296,970	$257,716	$39,254
Incremental cost per QALY gained			$30,907

Sensitivity Analysis

- Results were robust to changes in input parameter values and treatment scenarios (Figure 2, Table 7).
- For all ranges tested in the sensitivity analysis, the incremental cost per QALY gained remained below $50,000 (Figure 2).

Figure 2. One-Way Sensitivity Analysis: Tornado Diagram

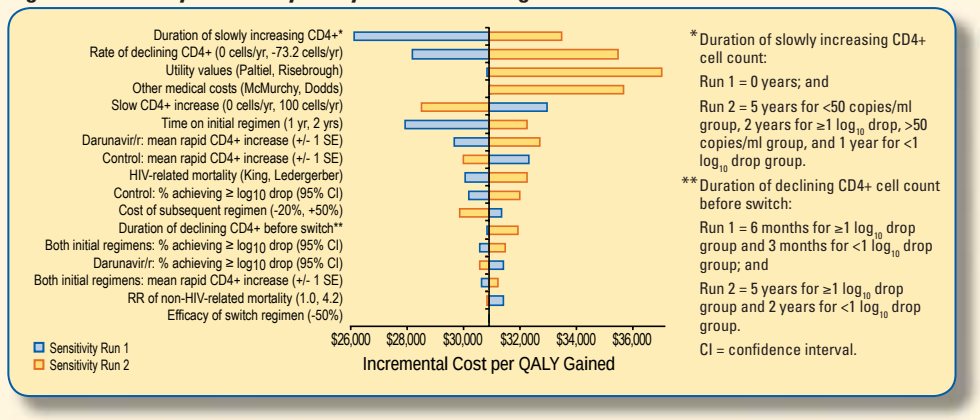
Duration of slowly increasing CD4+*
Rate of declining CD4+ (0 cells/yr, -73.2 cells/yr)
Utility values (Paltiel, Risebrough)
Other medical costs (McMurchy, Dodds)
Slow CD4+ increase (0 cells/yr, 100 cells/yr)
Time on initial regimen (1 yr, 2 yrs)
Darunavir/r: mean rapid CD4+ increase (+/- 1 SE)
Control: mean rapid CD4+ increase (+/- 1 SE)
HIV-related mortality (King, Ledergerber)
Control: % achieving ≥ log10 drop (95% CI)
Cost of subsequent regimen (-20%, +50%)
Duration of declining CD4+ before switch**
Both initial regimens: % achieving ≥ log10 drop (95% CI)
Darunavir/r: % achieving ≥ log10 drop (95% CI)
Both initial regimens: mean rapid CD4+ increase (+/- 1 SE)
RR of non-HIV-related mortality (1.0, 4.2)
Efficacy of switch regimen (-50%)
Sensitivity Run 1
Sensitivity Run 2
$26,000
$28,000
$30,000
$32,000
$34,000
$36,000
Incremental Cost per QALY Gained
*Duration of slowly increasing CD4+ cell count:
Run 1 = 0 years; and
Run 2 = 5 years for <50 copies/ml group, 2 years for ≥1 log10 drop, >50 copies/ml group, and 1 year for <1 log10 drop group.
**Duration of declining CD4+ cell count before switch:
Run 1 = 6 months for ≥1 log10 drop group and 3 months for <1 log10 drop group; and
Run 2 = 5 years for ≥1 log10 drop group and 2 years for <1 log10 drop group.
CI = confidence interval.

Table 7. Results of Variability Analyses

Scenarios	QALYs		Total Costs		Incremental Cost per QALY Gained
	Darunavir/r	Control	Darunavir/r	Control	
Base case[1]	8.05	6.78	$296,972	$257,717	$30,907
Time horizon					
5 years	3.62	3.39	$135,663	$127,600	$34,135
10 years	5.90	5.31	$216,106	$198,693	$29,320
British Columbia population age, gender and CD4+ distributions[2]					
British Columbia population	8.17	6.90	$300,574	$261,551	$30,708
Tipranavir use in first control regimen (switch regimen is POWER 1 and POWER 2 control regimen)					
0%	7.83	6.52	$263,676	$223,103	$30,927
20%	7.83	6.58	$263,676	$226,387	$29,733
50%	7.83	6.66	$263,676	$231,314	$27,719
100%	7.83	6.81	$263,676	$239,526	$23,604
British Columbia rate (22.2%)	7.83	6.58	$263,676	$226,749	$29,594
Enfuvirtide use in first darunavir/r and control regimens					
0%	7.99	6.74	$274,684	$245,621	$23,283
20%	8.01	6.76	$284,415	$251,313	$26,350
40%	8.04	6.77	$294,148	$257,145	$29,267
60%	8.06	6.79	$303,883	$263,118	$32,038
British Columbia rate (31.25%)	8.03	6.77	$289,890	$254,576	$28,009

[1]The base-case values for the variables changed in the scenario analysis are as follows: time horizon lifetime, gender distribution: 88.6% males, 11.4% females; age distribution: 20-39, 27.5%; 40-64, 70.6%; 65+, 2.0%; starting CD4+ cell-count distribution: 0-50, 23.1%; 51-100, 15.3%; 101-200, 24.7%; 201-350, 18.8%; 351-500, 9.8%; >500, 8.2%; tipranavir not used in first control regimen mix; and enfuvirtide use matches that in POWER 1 and POWER 2 clinical trials for darunavir/r and control and in RESIST 1 and RESIST 2 clinical trials for the switch regimen.

[2]British Columbia population based on analysis of British Columbia Centre for Excellence data. Gender distribution: 91.4% male, 8.6% female. Age distribution: 20-39 = 32.4%; 40-64 = 65.7%; >65 = 1.9%. CD4+ cell count distribution: 0-50 = 21.0%; 51-100 = 14.3%; 101-200 = 22.9%; 201-350 = 26.7%; 351-500 = 12.4%; >500 = 2.9%.

CONCLUSIONS

- When compared to current PIs, darunavir/r in combination with an OBR is cost-effective in treatment-experienced adults who have failed prior antiretroviral therapy.
- The model results were most influenced by assumptions about duration of efficacy, rate of decline in CD4+ cell count after virologic failure, utility values, and other medical care costs in each CD4+ cell-count range.
- Variations in practice patterns and population and model characteristics also influenced the results of the model.
- Nevertheless, darunavir/r remained cost-effective compared to standard of care over all the parameter ranges and variability factors tested.

REFERENCES

Cahn P, et al. Abstract PL14.3, 7th ICDTHI, Nov 14-18, 2004, Glasgow, UK.
Cook J, et al. *AIDS Research and Human Retroviruses* 1999; 15(6):499-508.
Cooper D et al. Abstract No. 561. 12th CROI; Feb 22-25, 2005; Boston, MA.
Data on file. Results of POWER 1 and POWER 2 clinical trials, 2006.
Deeks SG et al. *AIDS* 2002;16:201-7.
DHHS 2006; Panel on Antiretroviral Guidelines for Adults and Adolescents. http://AIDSinfo.nih.gov.
Dodds C et al. 2001; www.gpiatlantic.org/pdf/health/costofaids.pdf.
Garcia Fet al. *J Acquir Immune Defic Syndr* 2004;36(2):702-13.
Hicks et al. Abstract 3726, 44th ICAAC, Oct 31-Nov 2, 2004, Washington, D.C.
Hunt PW et al. *AIDS* 2003; 17(13):1907-15.
Jensen-Fangel S et al. *AIDS* 2004;18(1):89-97.
Katlama C et al. Abstract No. 520. 13th CROI; Feb 6-9, 2006; Denver, CO.
Kaufmann GR et al. *Arch Intern Med* 2003;163(18):2187-95.
King JT Jr et al. *Med Decis Making* 2003;23(1):9-20.
Krentz HB et al. *CMAJ* 2003;169(2):106-10.
Ledergerber B, et al. *The Lancet* 2004; 364(9428):51-62.
McMurchy D et al. *AIDS Care*. Ottawa: Health Canada; 1998.
Mocroft A et al. The Lancet 2003; 362 (9377):22-29.
Ontario Drug Benefit Program Formulary. 2006. http://www.health.gov.on.ca/english/providers/program/drugs/odbf_mn.html.
Paltiel AD, et al. Med Dec Mak 1998; 18(2 Suppl):S93-105.
Quebec Formulary. 2006. http://www.ramq.gouv.qc.ca/fr/professionnels/listmed/lm_tdmf.shtml.
Risebrough N et al. Abstract No. 103. 6th CROI; Jan 31-Feb 4, 1999; Chicago, IL.
Simpson KN et al. *HIV Clin Trials* 2004;5(5):294-304.
Smith CJ et al. AIDS 2003;17(7):963-9.
Statistics Canada. *The consumer price index*, June 2006. http://www.statcan.ca/bsolc/english/bsolc?catno=62-001-X&CHROPG=1.
Statistics Canada. *Deaths*, 2003. Ottawa: Minster of Industry; 2005. Catalogue No. 84F0211XIE. http://www.statcan.ca/cgi-bin/downpub/listpub.cgi?catno=84F0211XIE200300.
Tarwater PM et al. *J Acquir Immune Defic Syndr* 2001;27(2):168-75.