A Q-TWiST Analysis of Lapatinib Plus Letrozole Compared With Letrozole Alone as First-Line Therapy in Hormone Receptor Positive (HR+) Metastatic Breast Cancer (MBC)

Beth Sherrill,[1] Mayur Amonkar,[2] Bintu Sherif,[1] Julie Maltzman,[2] Lisa O'Rourke,[2] Stephen Johnston[3]

[1]RTI Health Solutions, Research Triangle Park, NC, United States;
[2]GlaxoSmithKline Oncology, Collegeville, PA, United States;
[3]Royal Marsden NHS Foundation Trust and Institute of Cancer Research, London, United Kingdom

BACKGROUND

- The objective of this study was to compare the quality-adjusted survival experience among patients with ErbB2-positive (HER2+) advanced or metastatic breast cancer (MBC) treated with either lapatinib plus letrozole (L+Let) or letrozole plus placebo (Let).
- Data Source:
 - The study was a phase 3, randomized, double-blind, multicenter trial comparing L+Let with Let.
 - The study included postmenopausal women with hormone receptor positive (HR+) (estrogen receptor-positive [ER+] and/or progesterone receptor-positive [PgR+]) advanced or MBC, who had not received previous therapy for advanced or metastatic disease.
 - Patients were randomized to receive either Let (2.5 mg once daily [QD]) with L (1,500 mg QD) or Let (2.5 mg QD) with a matching placebo.
 - The analyses presented here use data through June 3, 2008, data lock date.
- The quality-adjusted time without symptoms or toxicity (Q-TWiST) method was used to compare the tradeoff between toxicities (TOX) and delayed progression in a prospectively defined subset of the randomized intent-to-treat (ITT) population that overexpressed HER2.

METHODS

- In the Q-TWiST approach, the survival period is demarcated into health states representing varying levels of utility for patients.[1]
- First, overall survival (OS) was calculated for each treatment group using the product limit method. Curves for progression-free survival (PFS) and for time with TOX were overlaid onto the OS curve.
- Areas between the curves represent mean times in each health state, as defined in Table 1.

Table 1. Definitions for Health States

Health State	Definition
TOX	• Time after randomization with grade 3/4 AEs before progression • Truncated if AE duration was greater or equal to time to progression (i.e., censored as of progression or censoring for progression) • Zero if no qualifying AEs prior to disease progression
TWiST	• Remaining time after TOX before progression • Derived as the difference between mean PFS and mean TOX, based on Kaplan-Meier estimates • Censored at the time of the last independently assessed radiologic scan preceding the initiation of any alternative anticancer therapy
REL	• After progression, period until death from any cause or end of follow-up • Derived as the difference between mean OS and mean PFS, based on Kaplan-Meier estimates • Censored as of last available contact with patient or data lock date (June 3, 2008)

AE = adverse event; REL = relapse.

- The primary Q-TWiST analysis was performed on the prospectively defined HER2+ population from Study EGF30008, with TOX defined to include only grade 3/4 AEs. A sensitivity analysis was performed on the ITT population.
- The Q-TWiST score was calculated as follows:

$$\text{Q-TWiST} = (\mu TOX \times TOX) + (\mu TWIST \times TWiST) + (\mu REL \times REL)$$

where the multiplication of utility and time results in a quality-adjusted duration.

- A threshold utility analysis was carried out to determine combinations of utility weights under which Q-TWiST is statistically different between treatment groups. Treatment comparisons of Q-TWiST were made for a matrix of possible utility weight combinations where:
 - μTWIST held constant at 1
 - μTOX and μREL varied from 0 to 1 by 0.25, resulting in 25 combinations.
- Using this methodology, survival time is discounted under the assumption that days of sickness are of less use to a patient than days without sickness, resulting in a measure for quality-adjusted survival.

RESULTS

- Among 1,286 patients enrolled and randomized, 219 were identified as HER2+ (L+Let, n = 111; Let, n = 108).
- As of June 3, 2008, more than 50% of the HER2+ population was alive. Overall median follow-up for survival was approximately 140 weeks in the HER2+ population and 172 weeks in the ITT population.
- Table 2 shows the number of patients experiencing each event in the HER2+ subgroup.

Table 2. Patient Status by Treatment Group, HER2+ Population*

Status	L+Let (n = 111)	Let (n = 108)	Totals (N = 219)
AE (grade 3/4)	34	21	55
PFS event (progression or death due to any cause)	88	89	177
Deaths (all causes)	50	54	104

*AEs occurred prior to disease progression or censor date for PFS.

Figures 1 to 4 show survival curves.

Figure 1. Partitioned Survival Curve for Combination Therapy (L+Let) in HER2+ Subgroup (n = 111)*

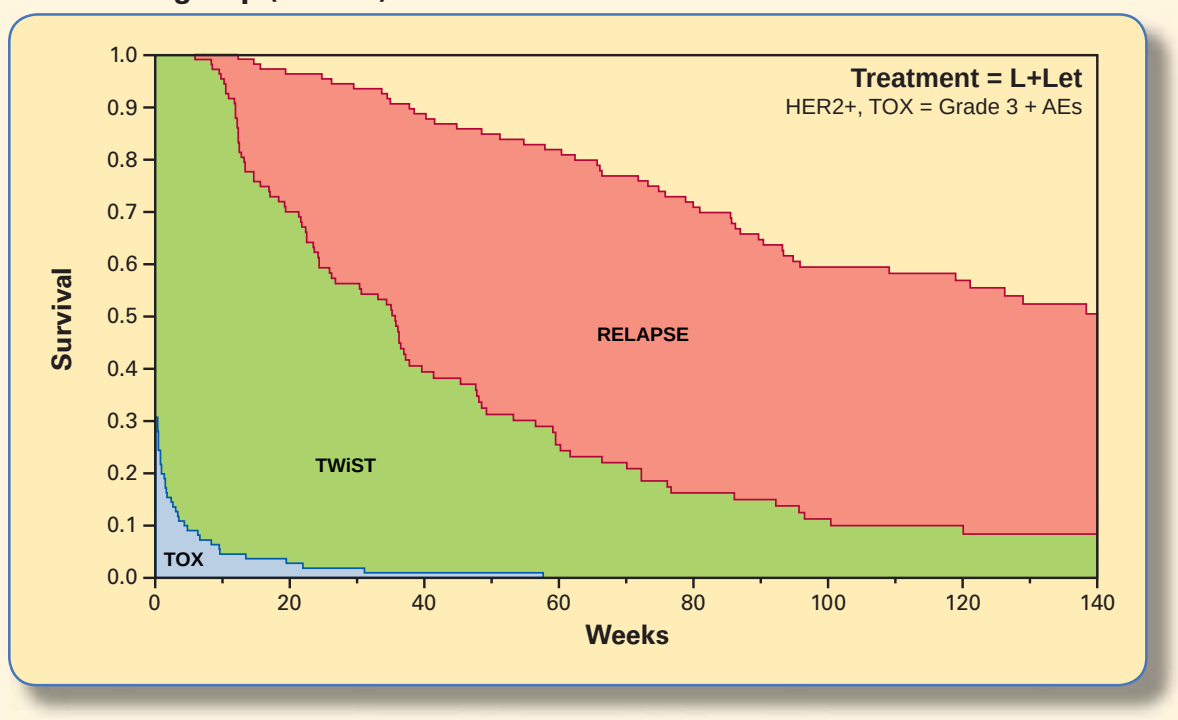
Treatment = L+Let
HER2+, TOX = Grade 3 + AEs
Survival
RELAPSE
TWiST
TOX
Weeks

Figure 2. Partitioned Survival Curve for Monotherapy (Let) in HER2+ Subgroup (n = 108)*

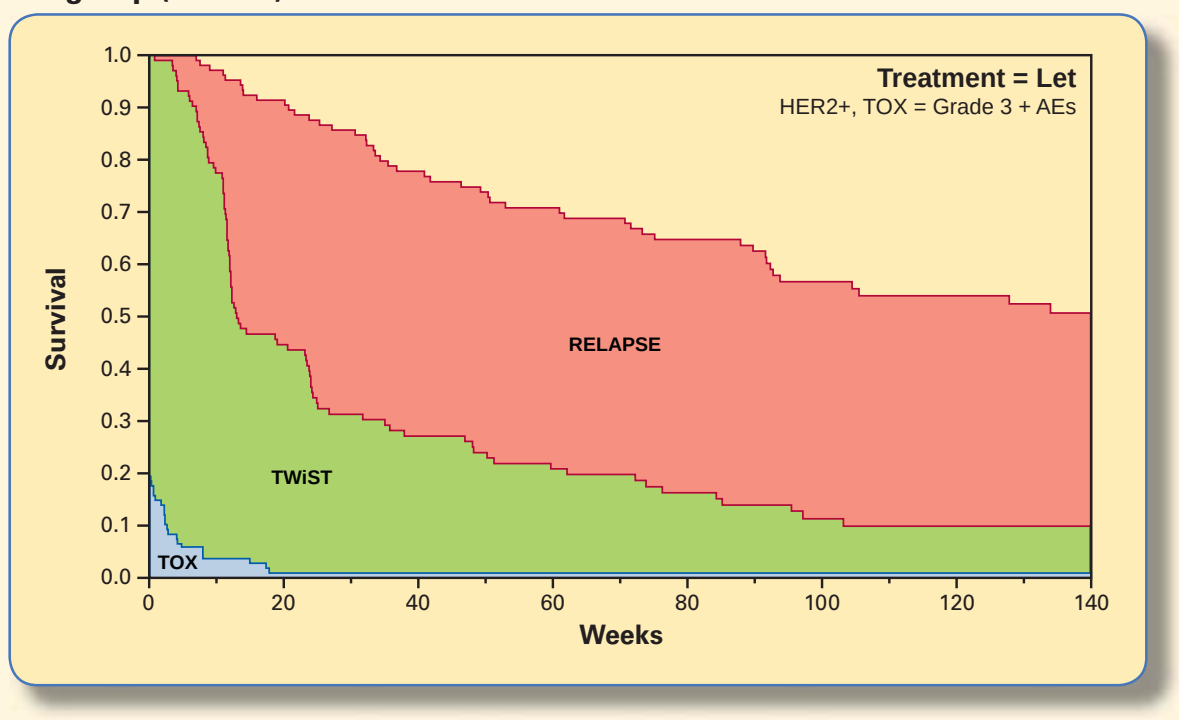
Treatment = Let
HER2+, TOX = Grade 3 + AEs
Survival
RELAPSE
TWiST
TOX
Weeks

Figure 3. Partitioned Survival Curve for Combination Therapy (L+Let) in ITT Population (n = 642)*

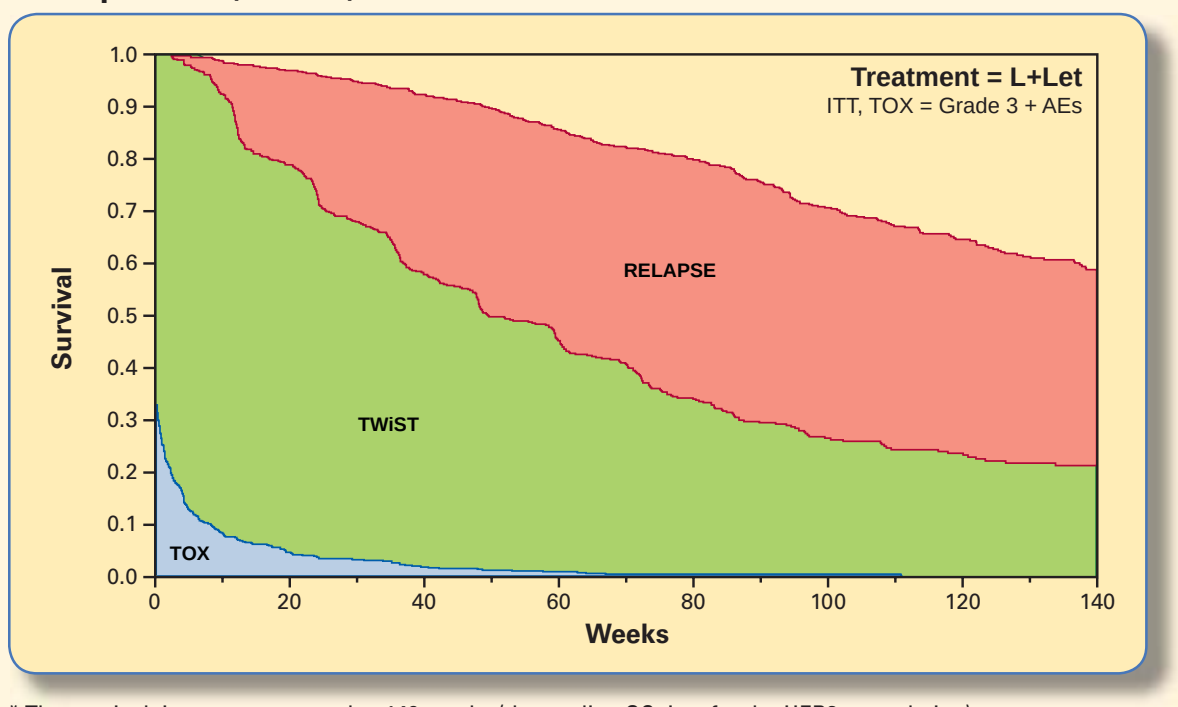
Treatment = L+Let
ITT, TOX = Grade 3 + AEs
Survival
RELAPSE
TWiST
TOX
Weeks

Figure 4. Partitioned Survival Curve for Monotherapy (Let) in ITT Population (n = 644)*

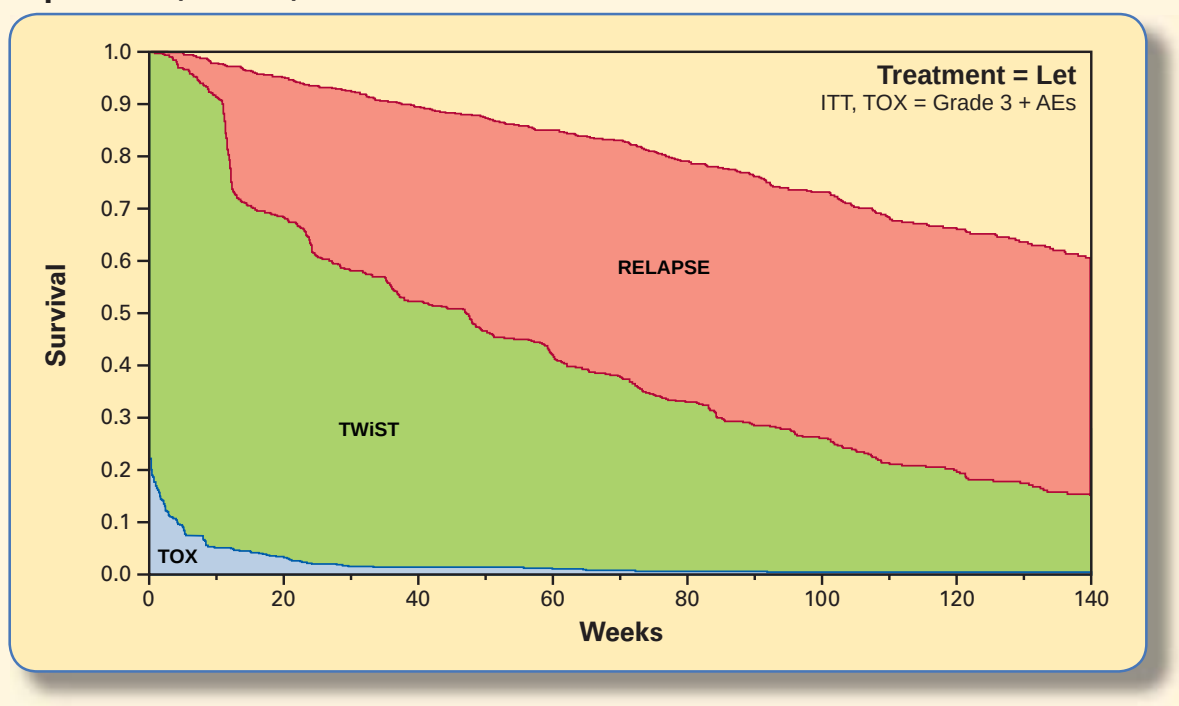
Treatment = Let
ITT, TOX = Grade 3 + AEs
Survival
RELAPSE
TWiST
TOX
Weeks

* The survival time was truncated to 140 weeks (the median OS time for the HER2+ population).

- Table 3 presents the unweighted mean durations of health states. There was no significant difference between groups in mean duration of serious AEs prior to progression in either the HER2+ subgroup or the ITT population.
- Using utility weights of 0.5 for both TOX and REL (i.e., counting 2 days of TOX or REL as 1 day of TWiST) resulted in a difference in quality-adjusted survival favoring L+Let of 8.8 weeks ($P = 0.09$) in the HER2+ subgroup.
- In the ITT population, the Q-TWiST difference at these utility levels was not statistically significant.

Table 3. Mean Duration of Health States (in Weeks)

Health State	L+Let	Let	Difference (L+Let) - Let	$P > \lvert Z \rvert$[a]
Primary Analysis (HER2+ Population)				
	(n = 111)	(n = 108)		
TOX: grade 3/4 AEs	1.95	2.14	-0.19	0.8959
TWiST	43.95	34.44	9.51	0.0973
RELAPSE	60.13	61.41	-1.28	0.8400
Q-TWiST[b]	74.99	66.22	8.77	0.0899
Sensitivity Analysis (ITT Population)				
	(n = 642)	(n = 644)		
TOX: grade 3/4 AEs	2.97	2.36	0.62	0.2937
TWiST	67.92	61.03	6.90	0.0407
RELAPSE	60.96	67.89	-6.93	0.0337
Q-TWiST[b]	99.89	96.15	3.74	0.1811

[a] Null hypothesis: Difference (L+Let) – Let = 0.
[b] Q-TWiST when uTOX = uREL = 0.5.

- Figure 5 illustrates the Q-TWiST differences across the entire matrix of hypothetical utility weights. Utility weights for REL and TOX are shown on the X and Y axes. The magnitude of the Q-TWiST difference (in weeks) is given by the numbered lines within each plot, with positive numbers favoring combination therapy over monotherapy. Shaded areas represent different levels of statistical significance; green areas depict utility weight combinations for which groups are statistically different ($P < 0.05$).
- In the HER2+ subgroup, the Q-TWiST difference between groups ranged from 8 to 9.5 weeks, favoring combination therapy for all hypothetical utility levels, but none of the comparisons were statistically significant at $P = 0.05$.
- In the ITT population, Q-TWiST differences ranged from 0 to 7.5 weeks, favoring combination therapy for all except one of the 25 utility combinations. Differences in favor of the combination therapy were statistically significant only when the utility for the relapse state was set to zero.

Figure 5. Contour Graphs Showing Q-TWiST Difference (in Weeks) Between Treatment (L+Let vs. Let) Varying Utility Levels*

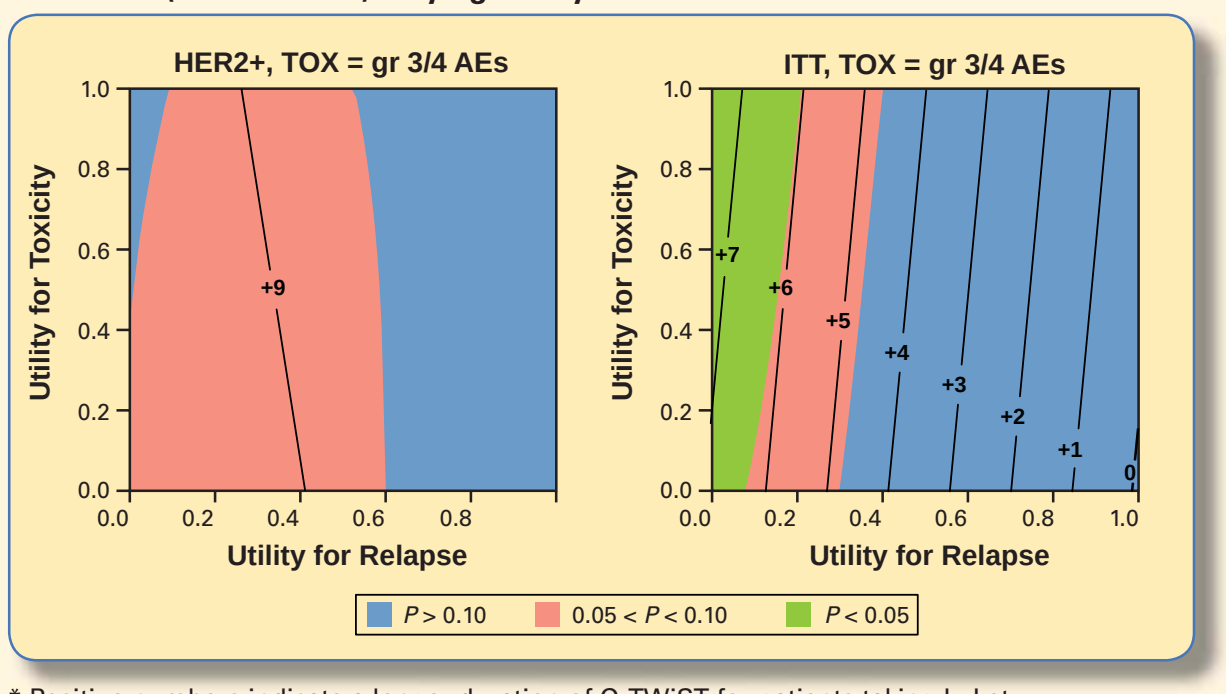
HER2+, TOX = gr 3/4 AEs
ITT, TOX = gr 3/4 AEs
Utility for Toxicity
Utility for Relapse
+9
+7
+6
+5
+4
+3
+2
+1
0
P > 0.10
0.05 < P < 0.10
P < 0.05

* Positive numbers indicate a longer duration of Q-TWiST for patients taking L+Let.

CONCLUSIONS

- Results are consistent with the Q-TWiST advantage previously reported for L combined with capecitabine versus monotherapy for women with previously treated HER2+ MBC.[2]
- The significantly longer PFS observed in HER2+, HR+ MBC patients taking the combination of L+Let versus Let was achieved without significant differences in mean duration of serious AEs.[3]
- Quality-adjusted survival was favored for the combination arm in the HER2+ and ITT populations.

REFERENCES

1. Glasziou PP, Simes RJ, Gelber RD. Quality adjusted survival analysis. Stat Med 1990;9:1259-76.
2. Sherrill B, Amonkar MM, Stein S, Walker M, Geyer C, Cameron D. Q-TWiST analysis of lapatinib combined with capecitabine for the treatment of metastatic breast cancer. Br J Cancer 2008;99:711-5.
3. Johnston S, Pegram M, Press M, Pippen J, Pivot X, Gomez H, et al. Lapatinib combined with letrozole vs. letrozole alone for front line postmenopausal hormone receptor positive (HR+) metastatic breast cancer (MBC): first results from the EGF30008 Trial. Abstract #46 presented at the 31st Annual San Antonio Breast Cancer Symposium Annual Meeting, San Antonio, TX. December 10-14, 2008.

CONTACT INFORMATION

Beth Sherrill, MStat
Global Head, Biometrics

RTI Health Solutions
200 Park Offices Drive
Research Triangle Park, NC 27709-2194

Phone: +1.919.541.8094
Fax: 919-541-7222
E-mail: bsherrill@rti.org

Presented at: The Joint 15th Congress of the European Cancer Organisation and 34th Congress of the European Society for Medical Oncology

September 20–24, 2009

Berlin, Germany