Quality of Life and Quality-Adjusted Survival (Q-TWiST) in Patients Receiving Lapatinib Plus Letrozole as First-Line Therapy in Hormone Receptor Positive, HER2+ Metastatic Breast Cancer

Beth Sherrill,[1] Bintu Sherif,[1] Mayur M. Amonkar,[2] Yun Wu,[1] Julie Maltzman,[2] Lisa O'Rourke,[2] Stephen Johnston[3]

[1]RTI Health Solutions, Research Triangle Park, NC, United States; [2]GlaxoSmithKline Oncology, Collegeville, PA, United States; [3]Royal Marsden NHS Foundation Trust & Institute of Cancer Research, London, United Kingdom

BACKGROUND

- The data source for this study is a phase 3, randomized, double-blind, multicenter trial comparing lapatinib plus letrozole (L+Let) with letrozole plus placebo (Let).
 - The study included postmenopausal women with hormone receptor positive (HR+) (estrogen receptor-positive [ER+] and/or progesterone receptor-positive [PgR+]) advanced or metastatic breast cancer (MBC), who had not received previous therapy for advanced or metastatic disease.
 - Patients were randomized to receive either Let (2.5 mg once daily [QD]) with L (1,500 mg QD) or Let (2.5 mg QD) with a matching placebo.
 - The analyses presented here were based on a prospectively defined *ErbB2*-positive (HER2+) population with data lock date June 3, 2008.
- The efficacy analysis of this clinical trial has revealed that progression-free survival (PFS) was significantly prolonged for L+Let compared with Let (8.2 vs. 3 months; hazard ratio (95% CI) = 0.71 (0.53, 0.96); *P* = 0.019) in the HER2+ population.[1]

OBJECTIVE

The objectives of these analyses were as follows:

- Evaluate and compare the two treatment arms with respect to change in quality of life (QOL).
- Use the quality-adjusted time without symptoms of disease or toxicity of treatment (Q-TWiST) method to compare the treatment toxicity and time-dependent clinical outcomes simultaneously.[2]

METHODS

QOL Assessments

- QOL was assessed using the Functional Assessment of Cancer Therapy-Breast (FACT-B) questionnaire (Version 4), which measures multidimensional QOL in patients with breast cancer over a recall period of 7 days.[3]
- FACT-B produces five subscale scores—physical well-being (PWB), social/family well-being (SWB), emotional well-being (EWB), functional well-being (FWB), and breast cancer subscale (BCS)—which are combined as follows:

FACT-B total score = PWB + SWB + EWB + FWB + BCS

FACT general (FACT-G) score = PWB + SWB + EWB + FWB

Trial outcome index (TOI) score = PWB + FWB + BCS

- Higher scores on the FACT-B scales indicate a higher QOL.
- A clinically meaningful change or minimum important difference (MID) has been estimated based on previous studies (2-3 points for the BCS, 7-8 points for the FACT-B total score, 5-6 points for the FACT-G and the TOI scores).[4]
- The FACT-B questionnaire was completed on day 1 predose, every 12 weeks, and at study withdrawal.
- All withdrawals were included in analyses up to the time of withdrawal. Analyses were based on observed data.
- Changes from baseline in the FACT-B total score, FACT-G score, and TOI score were analyzed in the HER2+ population using analysis of covariance with baseline value as a covariate.
- In a responder analysis, patients achieving MID in QOL scores (QOL responders) were compared using Fisher's exact test.

Q-TWiST Method

- The survival curves for each treatment arm were partitioned into three health states representing varying levels of utility for patients.
- First, overall survival (OS) was calculated for each treatment group using the product limit method. Curves for PFS and for time with toxicity (TOX) were overlaid onto the OS curve.
- Areas between the curves represent mean times in each health state, as defined in Figure 1.

Figure 1. Health States

TOX	TWiST	REL
Period after randomization, with grade 3/4 AEs prior to disease progression or censoring for progression	Period without toxicity or symptoms prior to disease progression	Period following disease progression and ending with death or censoring

AE = adverse event; REL = relapse.

- The Q-TWiST score was calculated as follows:

$$\text{Q-TWiST} = (\mu TOX \times TOX) + (TWiST) + (\mu REL \times REL)$$

 where the multiplication of utility and time results in quality-adjusted survival duration.
- A threshold utility analysis was carried out to determine combinations of utility weights under which Q-TWiST is statistically different between treatment groups. Treatment comparisons of Q-TWiST were made for a matrix of possible utility weight combinations where:
 - μTOX and μREL varied from 0 to 1 by 0.25, resulting in 25 combinations.
 - Per convention, TWiST was assigned a nominal utility value of 1.
- Using this methodology, survival time is discounted under the assumption that days of sickness are of less use to a patient than days without sickness, resulting in a measure for quality-adjusted survival.

QOL RESULTS

- Among 1,286 patients randomized, 219 were identified as HER2+ (L+Let, n = 111; Let, n = 108).
- Because QOL assessments were stopped after treatment termination, few patients completed the questionnaire after week 48, and the results reported here are only for the visits up to week 48.
- On average, patients in the two treatment arms had similar baseline values in all the FACT-B scores (Table 1).

Table 1. Summary of Baseline FACT-B Subscale Scores, FACT-B Total Scores, FACT G Scores, and TOI Scores by Treatment Arm (HER2+ Population)

Assessment	L+ Let (n = 111)		Let (n = 108)	
	n	Mean (SD)	n	Mean (SD)
PWB subscale (0-28)	106	21.8 (5.05)	99	21.2 (5.22)
SWB subscale (0-28)	109	20.9 (5.86)	98	22.4 (5.95)
EWB subscale (0-24)	110	15.6 (4.50)	100	16.0 (4.85)
FWB subscale (0-28)	110	17.5 (5.68)	100	17.7 (5.93)
BCS (0-36)	108	23.2 (5.19)	98	23.6 (5.98)
FACT-B total (0-144)	104	99.3 (19.16)	96	101.1 (19.31)
FACT-G (0-108)	105	75.9 (15.65)	98	77.4 (15.64)
TOI (0-92)	103	62.5 (12.77)	97	62.4 (13.65)

SD = standard deviation.

- The mean changes in subscale and total QOL scores were generally stable over time in both treatment arms for patients who stayed in the study (Figure 2), with no significant differences between groups.

Figure 2. Adjusted[a] Mean Change From Baseline for FACT-B Total Scores[b, c]

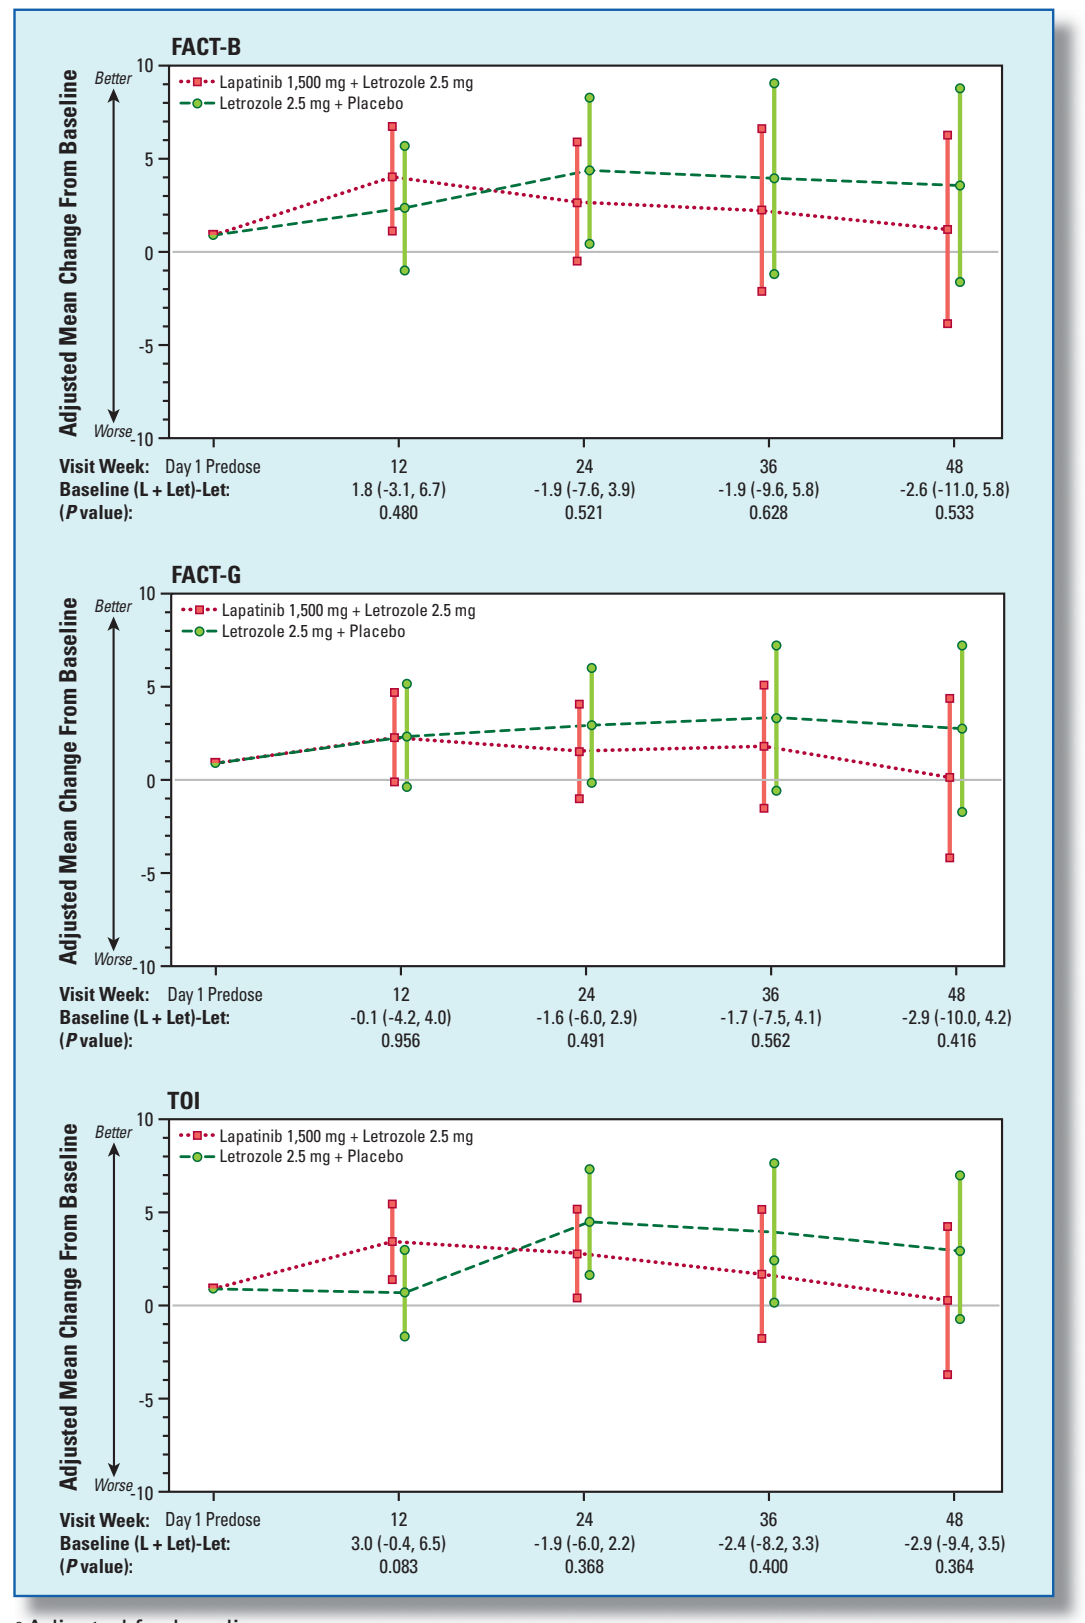
FACT-B
Lapatinib 1,500 mg + Letrozole 2.5 mg
Letrozole 2.5 mg + Placebo
Adjusted Mean Change From Baseline
Better
Worse
Visit Week: Day 1 Predose 12 24 36 48
Baseline (L + Let)-Let: 1.8 (-3.1, 6.7) -1.9 (-7.6, 3.9) -1.9 (-9.6, 5.8) -2.6 (-11.0, 5.8)
(P value): 0.480 0.521 0.628 0.533
FACT-G
Baseline (L + Let)-Let: -0.1 (-4.2, 4.0) -1.6 (-6.0, 2.9) -1.7 (-7.5, 4.1) -2.9 (-10.0, 4.2)
(P value): 0.956 0.491 0.562 0.416
TOI
Baseline (L + Let)-Let: 3.0 (-0.4, 6.5) -1.9 (-6.0, 2.2) -2.4 (-8.2, 3.3) -2.9 (-9.4, 3.5)
(P value): 0.083 0.368 0.400 0.364

[a] Adjusted for baseline score.
[b] The bars indicate ± 1.96 standard error.
[c] The analysis was performed based on observed data.

- In both treatment arms, 30% to 40% of patients had minimally important improvements in QOL during the study (Table 2) with no significant differences between groups.

Table 2. Summary of Comparison of QOL Response

QOL Score		L+ Let	Let	*P* Value for Treatment Difference[a]
FACT-B total	n[b]	98	85	
	≥ 8 (MID upper bound) ≥ 7 (MID lower bound)	33 (34%) 36 (37%)	29 (34%) 29 (34%)	> 0.99 0.76
FACT-G	n[b]	99	87	
	≥ 6 (MID upper bound) ≥ 5 (MID lower bound)	38 (38%) 41 (41%)	29 (33%) 34 (39%)	0.54 0.77
TOI	n[b]	97	87	
	≥ 6 (MID upper bound) ≥ 5 (MID lower bound)	33 (34%) 36 (37%)	29 (33%) 30 (34%)	> 0.99 0.76

[a] *P* values are from Fisher's exact test.
[b] n is number of subjects with baseline and at least one postbaseline score.

Q-TWIST RESULTS

- As of June 3, 2008, more than 50% of the HER2+ population was alive. Overall median follow-up for survival was 140 weeks.
- Table 3 presents the unweighted mean durations of health states. There was no significant difference between groups in mean duration of serious AEs prior to progression in the HER2+ population.

Table 3. Mean Duration of Health States (in Weeks)

Health State	L+Let (n = 111)	Let (n = 108)	Difference (L+Let) - Let	*P* > \| Z \|[a]
TOX: grade 3/4 AEs	1.95	2.14	-0.19	0.8959
TWiST	43.95	34.44	9.51	0.0973
REL	60.13	61.41	-1.28	0.8400
Q-TWiST[b]	74.99	66.22	8.77	0.0899

[a] Null hypothesis: Difference (L+Let) – Let = 0.
[b] Q-TWiST when $u_{TOX} = u_{REL} = 0.5$.

- Figure 3 and Figure 4 show partitioned survival curves, truncated to median OS time (140 weeks) with TOX defined to include only grade 3/4 AEs.

Figure 3. Partitioned Survival Curve for Combination Therapy (L+Let) in HER2+ patients (n = 111)

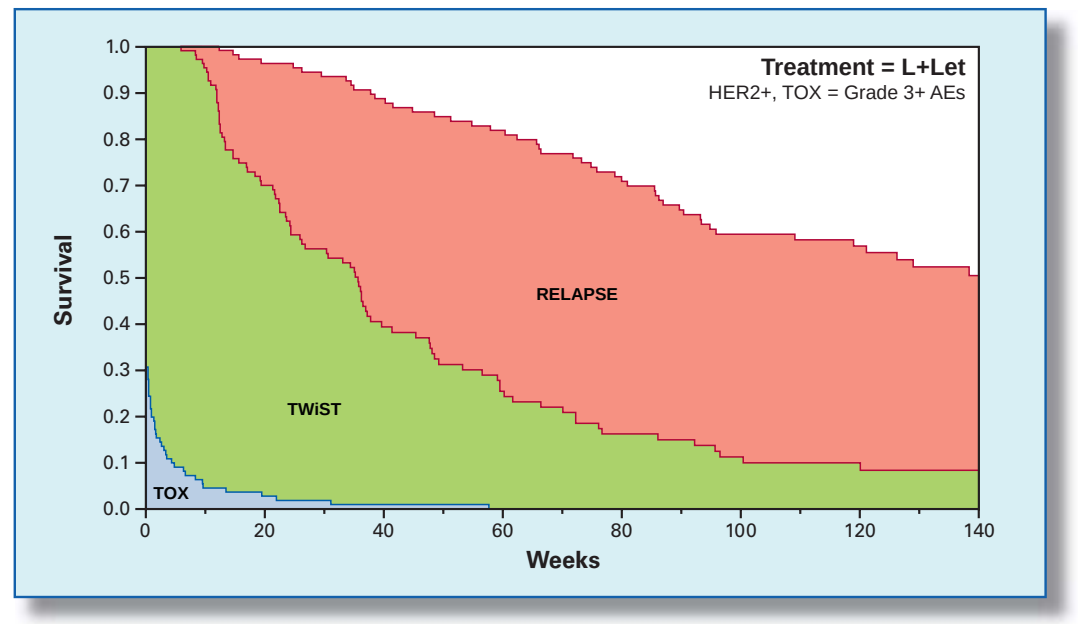
Treatment = L+Let
HER2+, TOX = Grade 3+ AEs
RELAPSE
TWiST
TOX
Survival
Weeks

Figure 4. Partitioned Survival Curve for Monotherapy (Let) in HER2+ patients (n = 108)

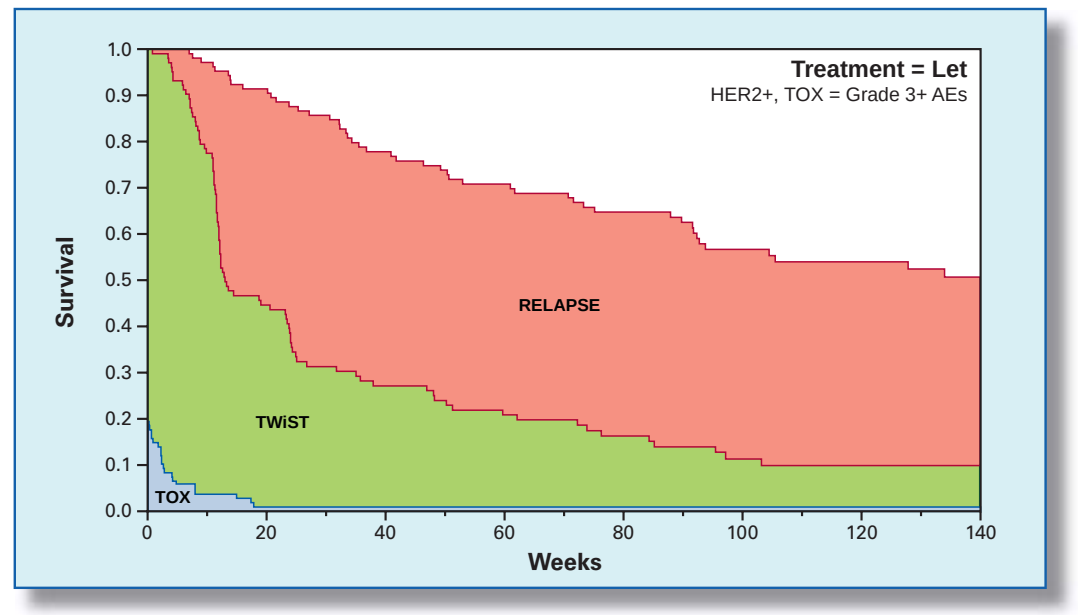
Treatment = Let
HER2+, TOX = Grade 3+ AEs
RELAPSE
TWiST
TOX
Survival
Weeks

- Figure 5 illustrates Q-TWiST differences of 9.5 weeks across the matrix of hypothetical utility weights, favoring combination therapy. Utility weights for REL and TOX are shown on the X and Y axes. The magnitude of the Q-TWiST difference (in weeks) is given by the numbered lines within each plot. Shaded areas represent different levels of statistical significance.
- A sensitivity analysis was performed using all AEs in the definition of the TOX state. In this scenario, there was less difference in QTWiST between the two treatment arms.

Figure 5. Contour Graphs Showing Q-TWiST Difference (in Weeks) Between Treatment (L+Let vs. Let) Varying Utility Levels[a]

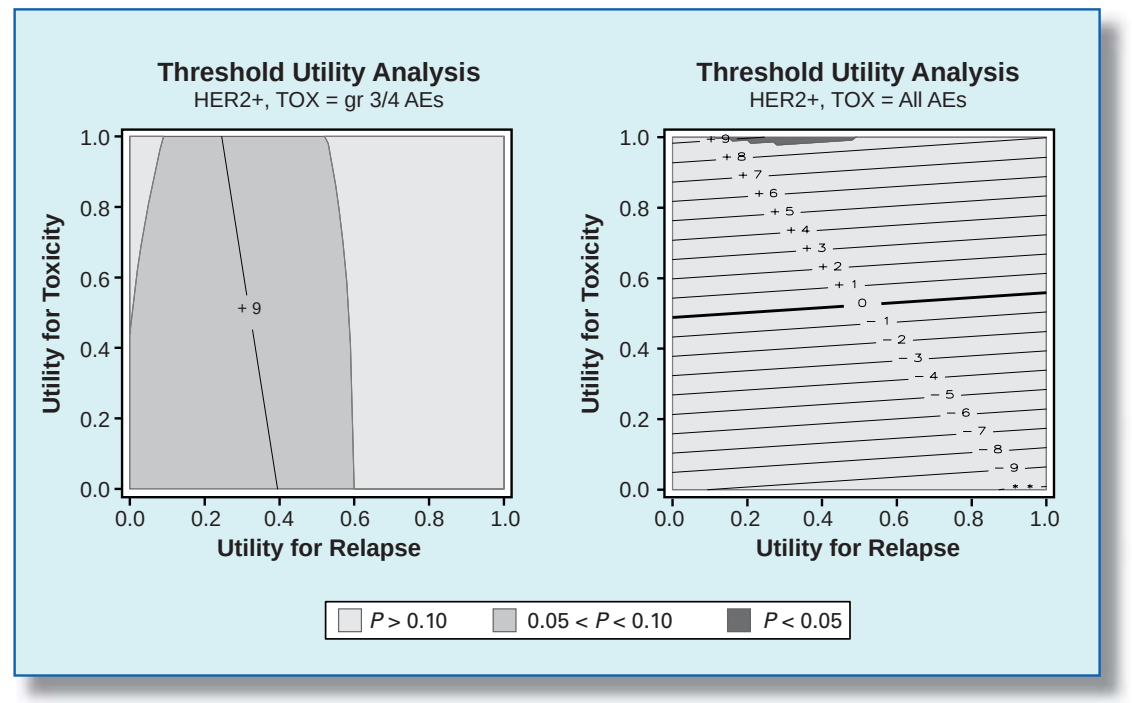
Threshold Utility Analysis
HER2+, TOX = gr 3/4 AEs
Threshold Utility Analysis
HER2+, TOX = All AEs
Utility for Toxicity
Utility for Relapse
P > 0.10
0.05 < P < 0.10
P < 0.05

[a] Positive numbers indicate a longer duration of Q-TWiST for patients taking L+Let.

CONCLUSIONS

- Subscale and total QOL scores were generally stable in both treatment arms for patients who stayed in the study, with approximately one-third of patients achieving improvements in QOL during treatment.
- The significantly longer PFS observed in patients taking the combination of L+Let versus Let was achieved without significant differences in mean duration of serious AEs.
- Quality-adjusted survival (QTWiST) was favored for the combination arm, but differences between groups were not statistically significant at hypothetical utility levels.
- Among HR+, HER2+ MBC patients, treatment with L+Let, a chemotherapy-free regimen, increased PFS while maintaining QOL and showing greater quality-adjusted survival when compared with Let alone.

REFERENCES

1. Johnston S, Pegram M, Press M, Pippen J, Pivot X, Gomez H, et al. Lapatinib combined with letrozole vs. letrozole alone for front line postmenopausal hormone receptor positive (HR+) metastatic breast cancer (MBC): first results from the EGF30008 Trial. Abstract #46 presented at the 31st Annual San Antonio Breast Cancer Symposium Annual Meeting, San Antonio, TX. December 10-14, 2008.
2. Glasziou PP, Simes RJ, Gelber RD. Quality adjusted survival analysis. Stat Med 1990;9:1259-76.
3. Brady MJ, Cella DF, Mo F, Bonomi AE, Tulsky DS, Lloyd SR, et al. Reliability and validity of the Functional Assessment of Cancer Therapy-Breast quality-of-life instrument. J Clin Oncol 1997;15:974-86.
4. Eton DT, Cella D, Yost KJ, Yount SE, Peterman AH, Neuberg DS, et al. A combination of distribution- and anchor-based approaches determined minimally important differences (MIDs) for four endpoints in a breast cancer scale. J Clin Epidemiol 2004;57:898-910.

CONTACT INFORMATION

Beth Sherrill, MStat
Global Head, Biometrics

RTI Health Solutions
200 Park Offices Drive
Research Triangle Park, NC 27709

Telephone: +1.919.541.1233
Fax: +1.919.541.7222
E-mail: bsherrill@rti.org

Presented at: 2009 Breast Cancer Symposium
October 8-10, 2009
San Francisco, CA, United States