MDS Poster NEW:Layout 1 13/6/08 17:01 Page 1

1058

Adherence Among Patients with Parkinson's Disease Taking Ropinirole Immediate Release At Least Three Times Daily

Heather M. Edin, MSPH[1]; Dana B. DiBenedetti, PhD[2]; Cheryl D. Hill, PhD[2]

[1]GlaxoSmithKline, Research Triangle Park, NC, USA; [2]RTI Health Solutions, Research Triangle Park, NC, USA

Introduction

Medication non-adherence is a serious problem facing current medical practice,[1,2] and research shows that patients who have a high frequency treatment regimen tend to be less compliant than patients who have a treatment regimen of lower frequency.[3] While adherence to medication has been demonstrated across a range of therapeutic areas, such data in Parkinson's disease (PD) are limited. The purpose of this study was to assess self-reported adherence, as well as drivers and consequences of non-adherence among patients with PD taking ropinirole immediate release (IR) at least three times daily (t.i.d.).

Methods

Study Design and Patients

- This survey was carried out in the USA between April and May 2006, in patients with PD who had been taking stable doses of ropinirole IR for at least 4 weeks.
- The survey consisted of 25 items to help identify patients' adherence and experiences with ropinirole IR, in addition to their reasons for non-adherence, the consequences of non-adherence, and their interest in a once-daily formulation. Patients were asked to respond to each question from a multiple choice of responses.
- Patients were eligible for inclusion if they were:
 - aged 18 years or older
 - had a diagnosis of idiopathic PD
 - taking a stable dose of ropinirole IR at least t.i.d. for a minimum of 4 weeks.
- All patients were recruited by TNS Healthcare GmbH through neurologists' offices, based on the inclusion criteria listed above. Physicians were responsible for identifying and recruiting patients who met the eligibility criteria by reviewing patient records at the beginning of the study.
- Eligible patients were contacted by mail or in person by their neurologist. After providing informed written consent, participating patients completed the survey at their physician's office and their neurologist completed a case report form to document patient and treatment history, and current dosing schedules.

Statistical Analysis

- Descriptive statistics were computed to summarize the responses to all survey items.
- Some items were collapsed into dichotomous categories and chi-square tests were used to test the association between adherence and other variables.
- Patients were defined as being adherent if they indicated that they never missed a dose of ropinirole IR, even if they reported late doses.
- No imputations were made for missing data and statistical inferences were based on the assumption of data being missing at random.

Results

Patients

- A total of 250 patients completed the survey.
- Patients were primarily white men with a mean (SD) age of 66.9 (10.5) years (**Table 1**).
- The most common dosing regime was t.i.d. (80% of patients) with a mean daily dose of 6.9 mg.

Table 1. Patient demographics and baseline characteristics

	N=250
Current age, years	
Mean (SD)	66.9 (10.5)
Age at first diagnosis, years	
Mean (SD)	61.0 (10.2)
Proportion of men, n (%)	156 (62)
Race/Ethnicity, n (%)	
White/Caucasian	226 (90)
Black	11 (4)
Hispanic	8 (3)
Asian	4 (2)
Hoehn and Yahr stage, n (%)	
0	1 (<1)
I	54 (22)
II	122 (49)
III	55 (22)
IV	15 (6)
V	3 (1)
Proportion of patients on PD medication (%)*	
Levodopa	55
Entacapone	22
Amantadine	16
Stalevo	7
None	36
Current employment status, n (%)	
Retired	150 (60)
Employed full time	49 (20)

*Total percentage may exceed 100%, as physicians were able to check more than one response.
PD, Parkinson's disease; SD, standard deviation.

- Participating patients had been taking ropinirole IR for a mean time of 32 months (range 1–120 months) and had been on their current ropinirole IR regimen for a mean time of 11.9 months (range 1–60).
- Most patients were either on ropinirole monotherapy (36%) or concomitant L-dopa (55%).
- Cardiovascular and gastrointestinal disorders were the most commonly occurring comorbidities, followed by depression.
- All patients except one reported having some type of health insurance, with more than 51% indicating that their primary health insurance was Medicare. Insurance coverage of prescription medications varied, with approximately 14% of the sample reporting full coverage, 74% reporting partial coverage with some co-payment, and 9% reporting no insurance coverage.

Self-Reported Adherence

- More than two-thirds (67%) of patients indicated that they had missed at least one dose of ropinirole IR and were classified as non-adherent. Specifically, in the last 7 days:
 - 24% of patients had missed at least one dose
 - 57% of patients were late with one or more doses.
- Adherence was unrelated to age, gender, employment status, education, dosage and frequency, current use of other PD medications, monotherapy versus combination therapy, dosing assistance strategies used, type of insurance and co-payment requirements, or percentage of "off" periods.

Reasons for Non-Adherence

- The most commonly reported reasons for non-adherence included forgetting to take treatment (44%), forgetting to take their treatment with them (35%), and being too busy to take treatment (31%) (**Figure 1**).
- In total, 44% of patients reported that taking ropinirole IR the prescribed number of times a day was 'very bothersome' or 'somewhat bothersome' rather than 'a little bothersome' or 'not bothersome at all'.
 - The most commonly reported reason for finding adherence bothersome was having too many pills to take each day (56%), followed by having to carry the medication around (40%), and having a complicated dosing schedule (37%) (**Figure 2**).

Figure 1. Most commonly reported reasons for non-adherence with ropinirole immediate release (N=250)

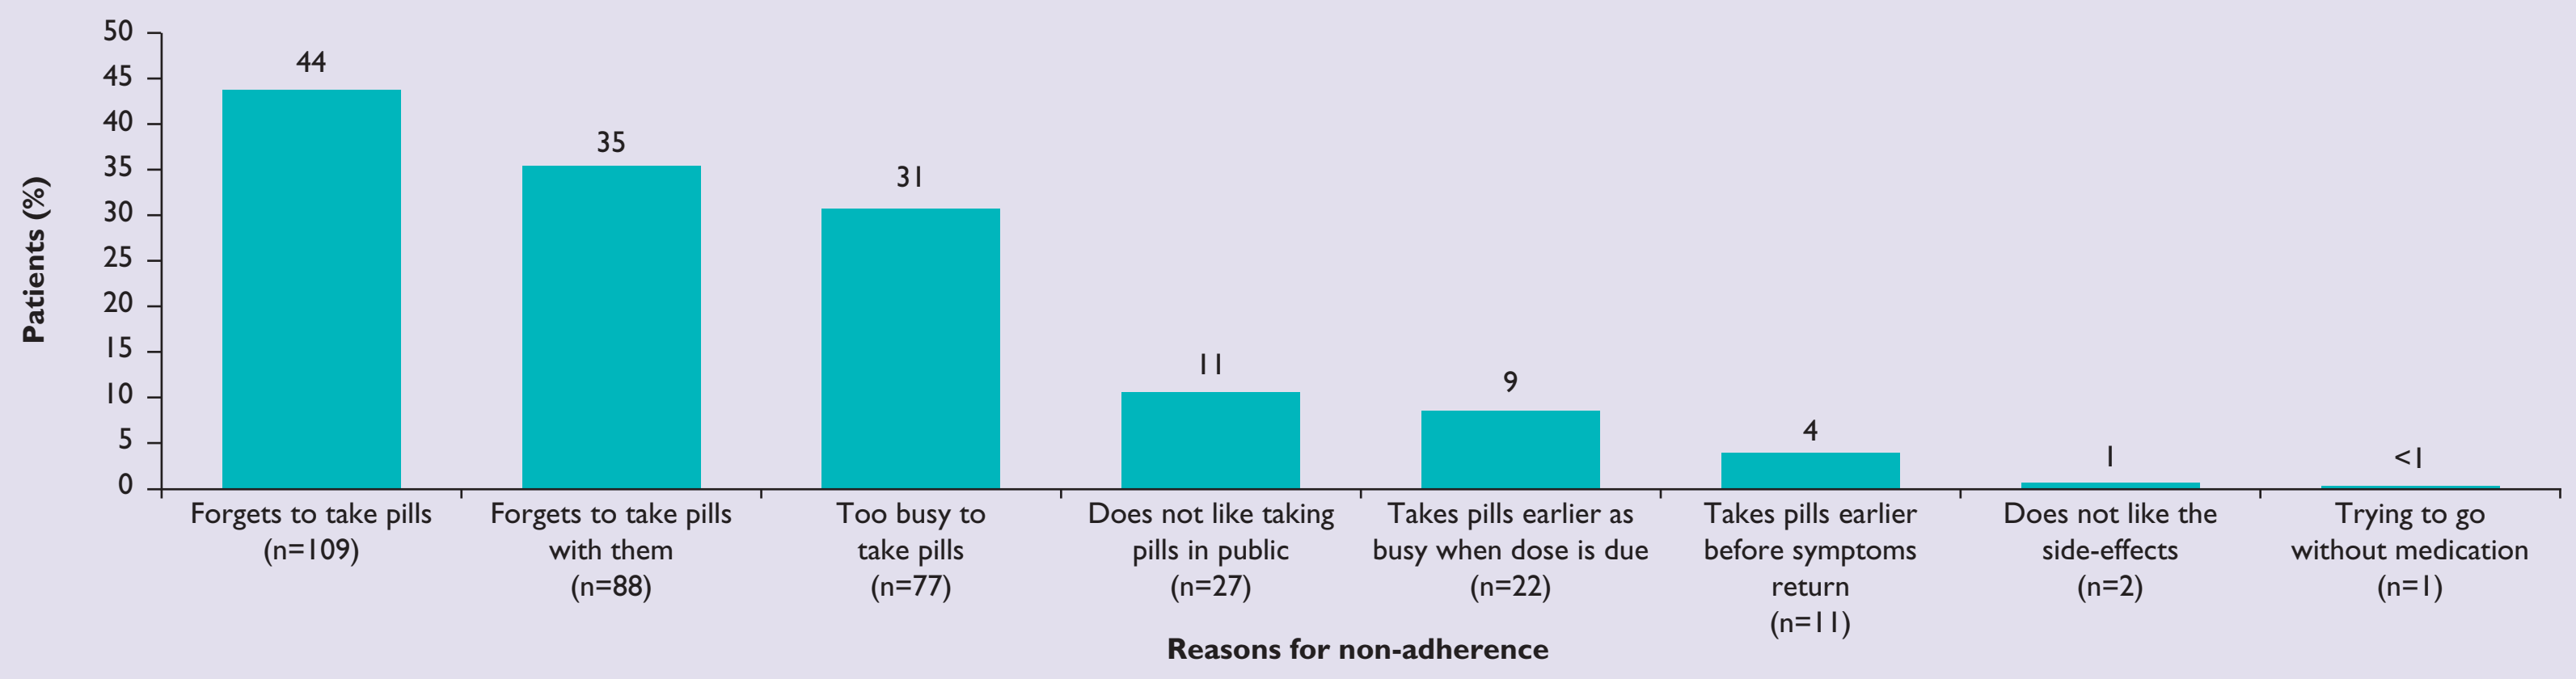
Patients (%)
50
45
40
35
30
25
20
15
10
5
0
44
35
31
11
9
4
1
<1
Forgets to take pills (n=109)
Forgets to take pills with them (n=88)
Too busy to take pills (n=77)
Does not like taking pills in public (n=27)
Takes pills earlier as busy when dose is due (n=22)
Takes pills earlier before symptoms return (n=11)
Does not like the side-effects (n=2)
Trying to go without medication (n=1)
Reasons for non-adherence

Total percentage exceeds 100% as participating patients may have provided more than one reason. Percentages have been rounded to the nearest whole number.

Figure 2. Reported reasons why taking ropinirole immediate release three times daily is bothersome

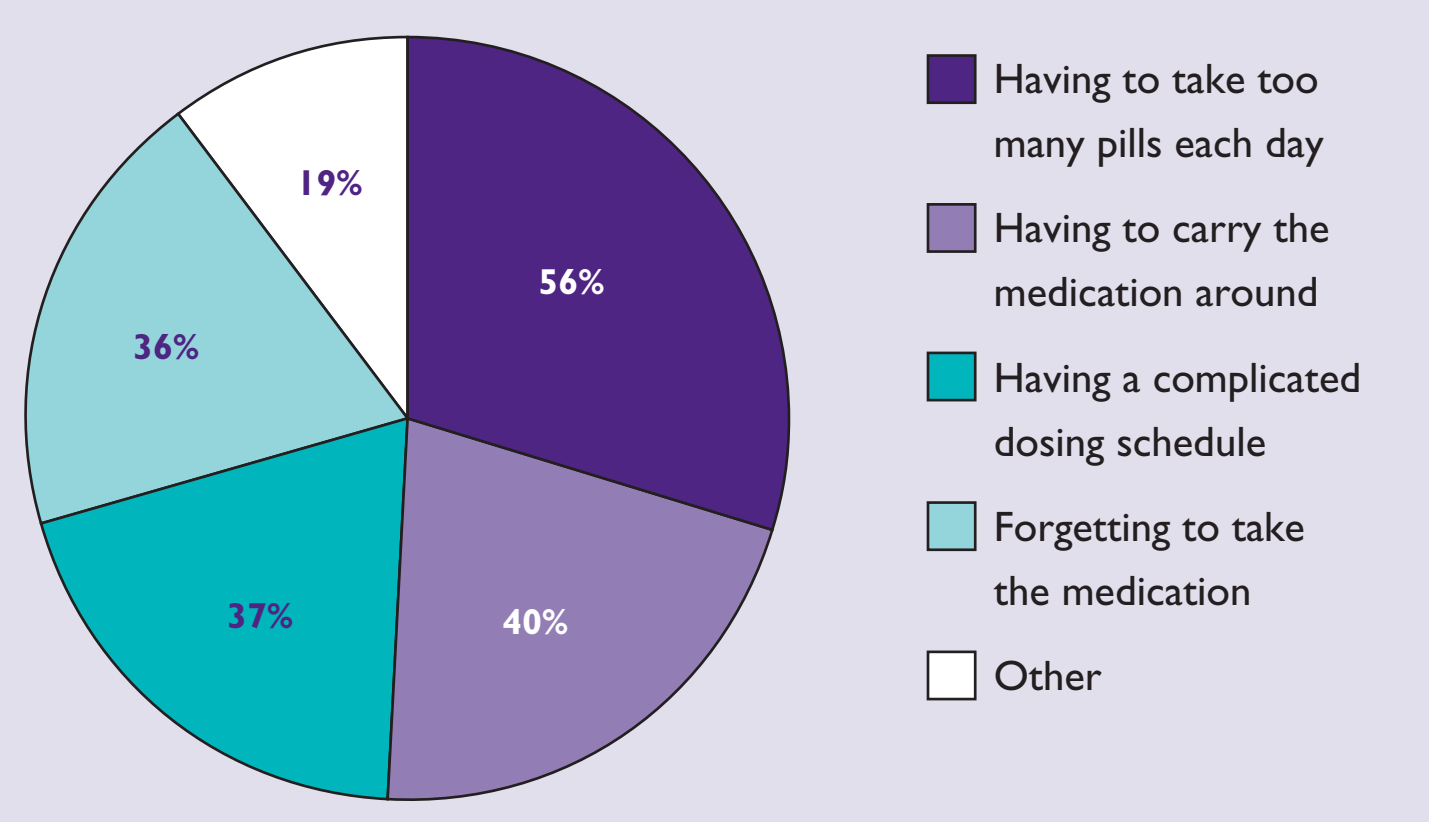
56%
40%
37%
36%
19%
Having to take too many pills each day
Having to carry the medication around
Having a complicated dosing schedule
Forgetting to take the medication
Other

Total percentage exceeds 100% as participating patients may have provided more than one reason.

Consequences of Non-Adherence

- The proportion of patients reporting a consequence related to missed or late doses of ropinirole IR was 75%.
 - The most commonly reported consequences of missed or late doses were an increase in tremors (43%), bradykinesia (34%), difficulty concentrating (25%), difficulty moving (24%), and difficulty walking (22%) (**Figure 3**).
 - The proportion of patients who reported development or a worsening of PD symptoms within 4 or fewer hours of a late or missed dose was 58%.
- Approximately half of the patient sample indicated that their current ropinirole IR dosing regimen interfered with their social activities (56%) and leisure activities (51%).
- Approximately 40% of patients reported interference with their work (outside the home and/or housework), sleep, mood, or concentration.
- Nearly one-third (30%) reported that their current ropinirole IR dosing regimen interfered with sexual activity.

Figure 3. Reported changes in Parkinson's disease symptoms experienced by patients when they miss or are late taking a dose of ropinirole immediate release (N=250)

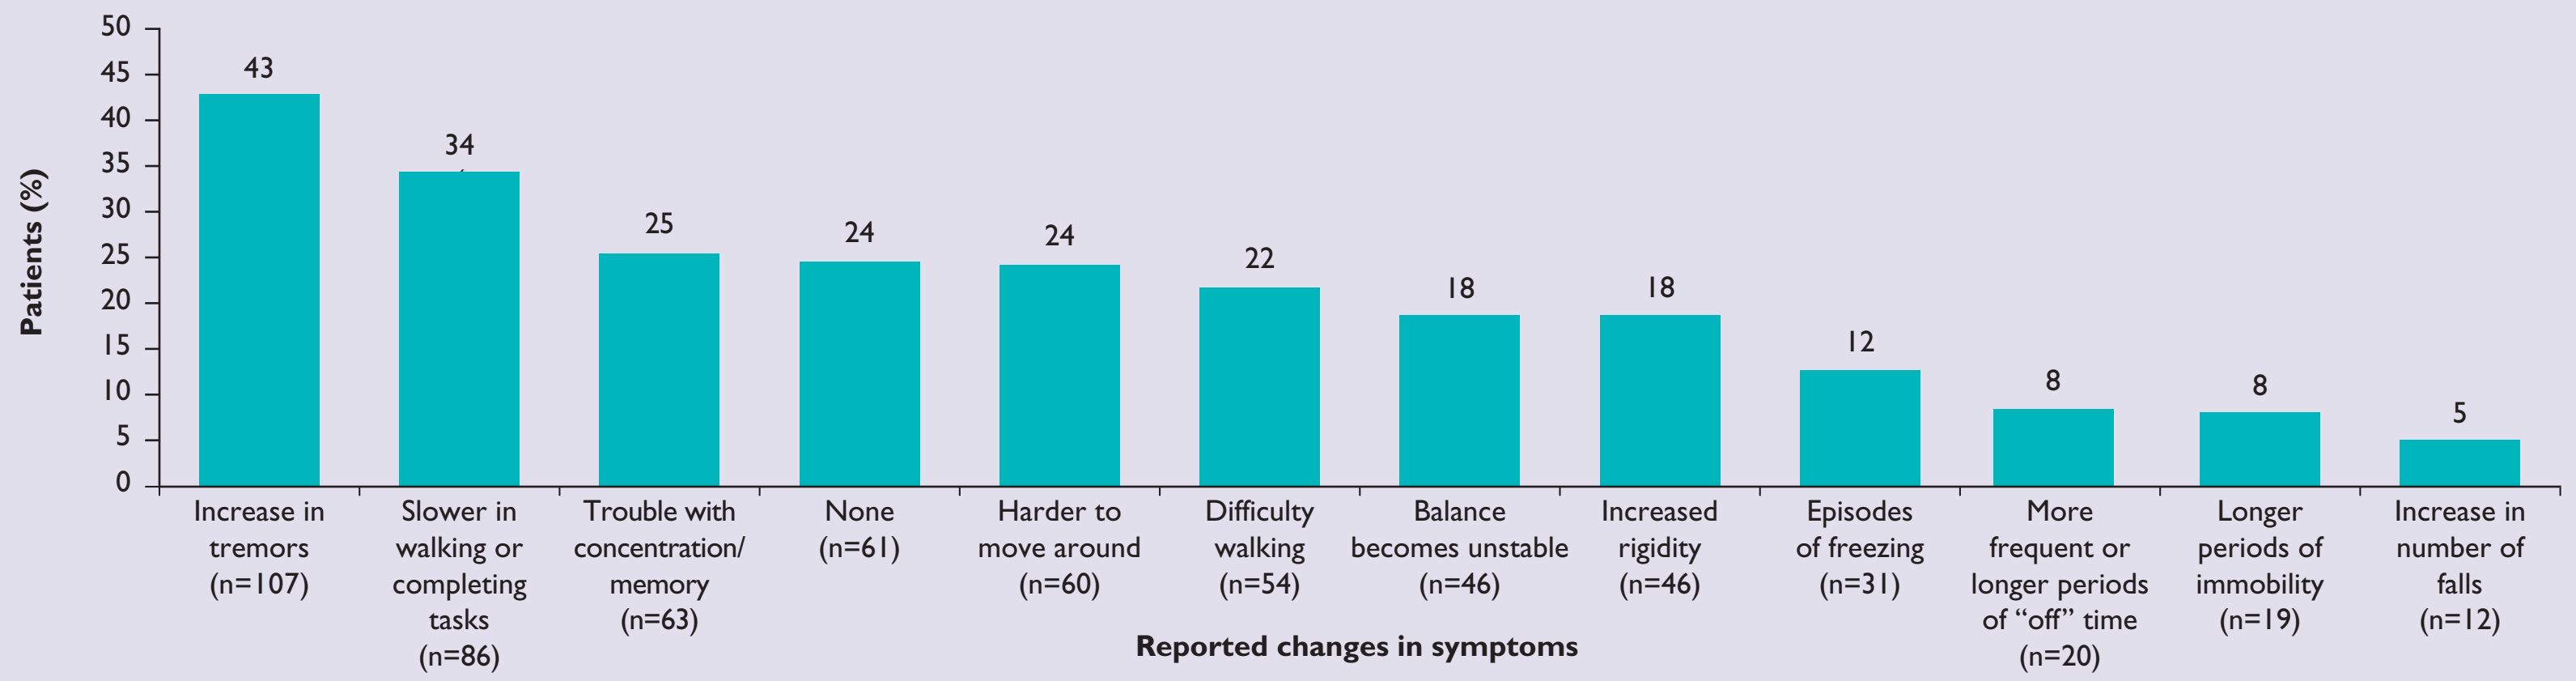
Patients (%)
50
45
40
35
30
25
20
15
10
5
0
43
34
25
24
24
22
18
18
12
8
8
5
Increase in tremors (n=107)
Slower in walking or completing tasks (n=86)
Trouble with concentration/ memory (n=63)
None (n=61)
Harder to move around (n=60)
Difficulty walking (n=54)
Balance becomes unstable (n=46)
Increased rigidity (n=46)
Episodes of freezing (n=31)
More frequent or longer periods of "off" time (n=20)
Longer periods of immobility (n=19)
Increase in number of falls (n=12)
Reported changes in symptoms

Total percentage exceeds 100% as participating patients may have provided more than one reason. Percentages have been rounded to the nearest whole number.

Patient Interest in Once-Daily Dosing

- When asked how interested they would be in a once-daily formulation of ropinirole from 'very interested' to 'very disinterested', the proportion of patients who reported being at least 'somewhat interested' was 88%.
 - The most common reasons for interest were: ease of remembering to take a once-daily formulation (86%); not having to carry medication around (31%); currently taking too many medications (17%); and being too busy to take medication more than once a day (14%).

Conclusions

- Pill burden and dosing schedule play an important role in the lack of adherence.
- Lack of adherence may contribute to morbidity and mortality and may be a driver behind repeat visits to the physician's office with unresolved symptom complaints and higher healthcare costs.
- Mechanisms to improve adherence to treatment regimens, including physician awareness, patient education, and improved dosing regimens may result in improved patient care and better outcomes.
- The majority of patients taking ropinirole IR in this survey were interested in the prospect of a once-daily dosing schedule.

References

1. O'Brien *et al. Med Care Rev* 1992;49:435–54.
2. Cramer *et al. Heart* 2002;88:203–6.
3. Claxton *et al. Clin Ther* 2001;23:1296–310.

Acknowledgments

This study was supported by GlaxoSmithKline. Patient recruitment and data collection were carried out by TNS Healthcare GmbH. Development of the patient survey and data analyses were carried out by RTI Health Solutions.

12th International Congress of Parkinson's Disease and Movement Disorders, Chicago, IL, USA, June 22–26, 2008